LSVP.23

DMD Patients Treated by SGT-001 Microdystrophin Gene Therapy Improve in the Objective Endpoint of Spontaneous Walking Velocity

Laurent Servais[1], J. Patrick Gonzalez[2], Carl A. Morris[2], Perry B. Shieh[3], Barry J. Byrne[4], Mélanie Annoussamy[5], Roxana Donisa Dreghici[2]
[1]University of Oxford, Oxford, UK; [2]Solid Biosciences, Charlestown, MA; [3]University of California at Los Angeles, Los Angeles, CA; [4]University of Florida College of Medicine, Powell Gene Therapy Center, Gainesville, FL; [5]SYSNAV, Vernon, France

Abstract

Duchenne muscular dystrophy (DMD) is a progressive, fatal neuromuscular disease characterized by the loss of muscle function beginning in early childhood. On average, patients experience declining ambulatory function until losing the ability to walk by early teens. Evaluation of ambulatory function is typically performed in the clinic using the 6MWT and NSAA, but these tools are prone to bias and variability that can make interpretation of longitudinal changes difficult. Stride velocity 95th centile (SV95C), which represents the fastest spontaneous strides in a patient's daily life as captured by a wearable device, is an objective assessment of peak performance accepted as a qualified secondary endpoint for DMD patients 5 years and older by the EMA and is currently evaluated in several clinical trials.

In the Phase 1/2 study IGNITE DMD, nine subjects were treated with SGT-001 microdystrophin gene therapy, with the first 3 receiving 5E13vg/kg and the next 6 administered 2E14vg/kg. Three untreated subjects were also enrolled in the study and analyzed as controls. SV95C was analyzed for all ambulatory subjects aged 5 and older at baseline. Control subjects showed diminished SV95C at all post-baseline timepoints up to 1 year, similar to natural history. SGT-001 treated subjects showed an average improvement in SV95C at all post-treatment timepoints, with positive results observed in both dose groups. Together with previously presented data, these results provide additional evidence that patients treated with SGT-001 may be experiencing a benefit to motor function and a clinical course that diverges from untreated patients.

Methods

SGT-001

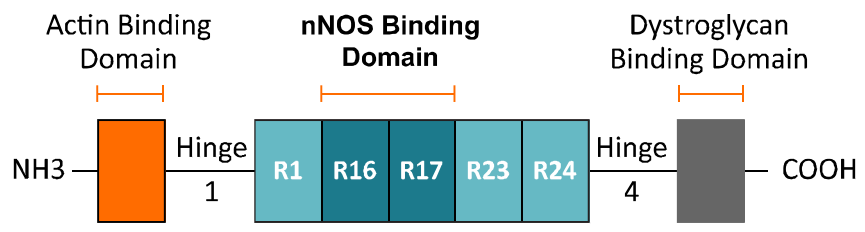
Actin Binding Domain
nNOS Binding Domain
Dystroglycan Binding Domain
NH3
Hinge 1
R1
R16
R17
R23
R24
Hinge 4
COOH

nNOS: neuronal nitric oxide synthase

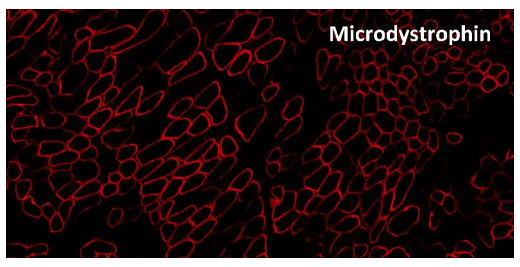
Microdystrophin

Biopsy from Pt. 5 at 18 months

- SGT-001 (rAAV9-CK8-h-µD5) is an AAV microdystrophin gene transfer therapy being evaluated for the treatment of DMD
- SGT-001 delivers a unique, rationally designed dystrophin surrogate to replace the absent protein in skeletal and cardiac muscles throughout the body

IGNITE DMD Study Design

- IGNITE DMD is a Phase I/II clinical trial to assess the safety and efficacy of SGT-001
 - Cohorts
 - n=3 subjects analyzed as controls
 - n=3 subjects at 5E13 vg/kg (Patients 1-3)
 - n=6 subjects at 2E14 vg/kg (Patients 4-9)
 - Primary Endpoints (Baseline to 1 Year)
 - Incidence of adverse events
 - Change in microdystrophin protein levels in muscle biopsies by Western blot
 - Select Secondary Endpoints
 - NSAA, 6MWT, PFTs, PROMs (PODCI), SV95C by ActiMyo
 - Additional evaluations performed at the 1.5-year timepoint and annually up to 5 years to assess long-term safety and efficacy
 - Enrollment in the study has concluded and subjects continue to undergo safety monitoring and efficacy evaluations

Stride Velocity 95th Centile (SV95C) Measurement

- Assessment of peak ambulatory performance in a real-world setting
- ActiMyo Wearable Device
 - A set of a wearable watch-like device and an associated docking unit that continuously records motion through specialized sensors that detect 3-dimensional movement
 - Sensors include tri-axial accelerometer, gyrometer, magnetometer, and barometer to measure linear acceleration angular velocity, magnetic field of movement and barometric altitude
 - Next generation device from SYSNAV: Syde
- SV95C as an Endpoint
 - 95th Percentile Stride Velocity: average speed (meters/second) of the 95th percentile of fastest strides taken
 - Serves as an indicator of maximal performance
 - Deemed an acceptable secondary endpoint in pivotal or exploratory drug therapeutic studies by the EMA when captured by a qualified wearable device (e.g. ActiMyo) worn at the ankle

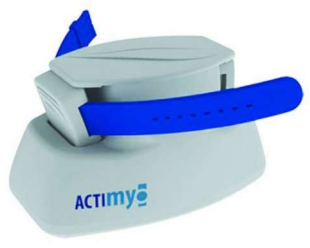
ACTImyo

ActiMyo

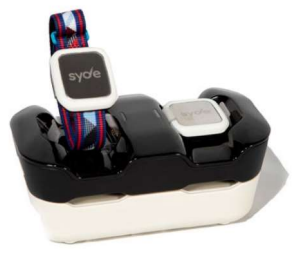
syde

Syde

Methods

Use of ActiMyo and SV95C Data Evaluation in IGNITE DMD

- ActiMyo devices worn by study subjects beginning at baseline prior to treatment
- Data generated in periods bracketing study visit timepoints compiled and analyzed to represent functional ability around that moment in time
- Subjects asked to wear devices up to the period bracketing the 1-year timepoint
 - Some subjects did wear devices longer, but total durations vary
- All data for ambulatory subjects 5 years or older achieving ≥50 hours of recording (minimum based on EMA qualification) in each defined visit window included in analysis
- Individual change from baseline calculated at post-treatment timepoints and expressed as a % change for inter-subject comparisons
- Data compared to a DMD natural history population of patients 5 years and older assembled in the EMA SV95C Endpoint Qualification Dossier by SYSNAV

Results

SGT-001 Treated Patients Show Improvements in SV95C at 1 Year Post-Dosing, Compared to Declines Observed in Control and Natural History Patients

Timepoint	Group	SV95C Change from Baseline (%)		
		n	Mean	SD
1 Year	DMD Natural History	28	-15.1	13.0
	Control	1	-17.2	N/A
	SGT-001 5E13 vg/kg	1	9.5	N/A
	SGT-001 2E14 vg/kg	5	8.8	13.4

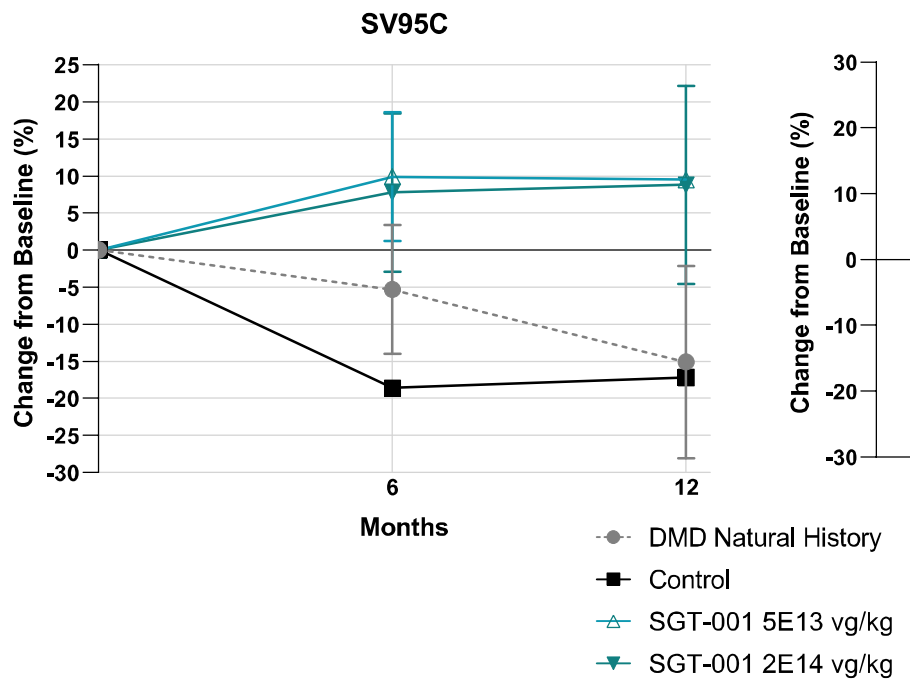
SV95C
Change from Baseline (%)
25
20
15
10
5
0
-5
-10
-15
-20
-25
-30
6
12
Months
DMD Natural History
Control
SGT-001 5E13 vg/kg
SGT-001 2E14 vg/kg

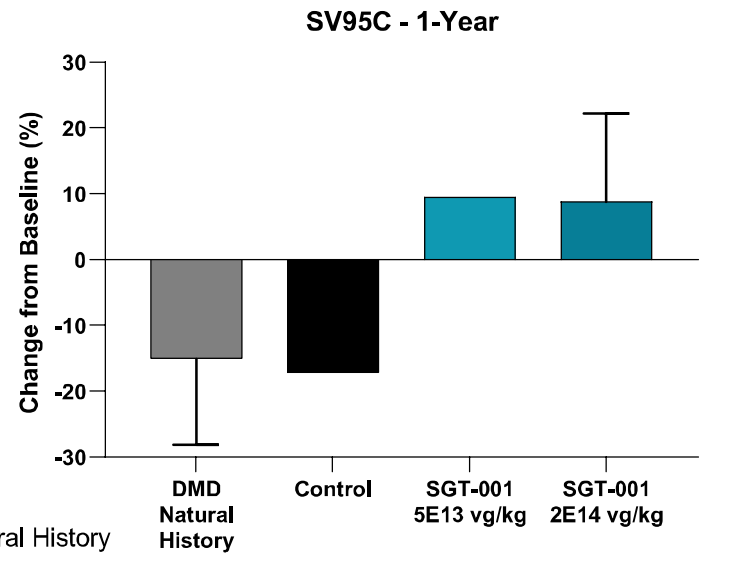
SV95C - 1-Year
Change from Baseline (%)
30
20
10
0
-10
-20
-30
DMD Natural History
Control
SGT-001 5E13 vg/kg
SGT-001 2E14 vg/kg

Improvements in SV95C are Consistent with Results from 6MWT and NSAA Clinical Evaluations in SGT-001 Treated Patients at 1 Year

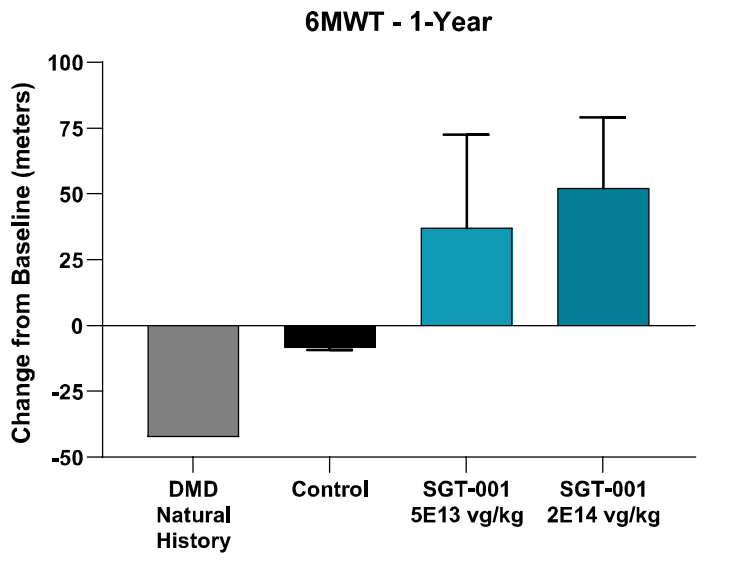
6MWT - 1-Year
Change from Baseline (meters)
100
75
50
25
0
-25
-50
DMD Natural History
Control
SGT-001 5E13 vg/kg
SGT-001 2E14 vg/kg

-42.3m expected decline in 12 months after age 7 (Mercuri et al 2016)

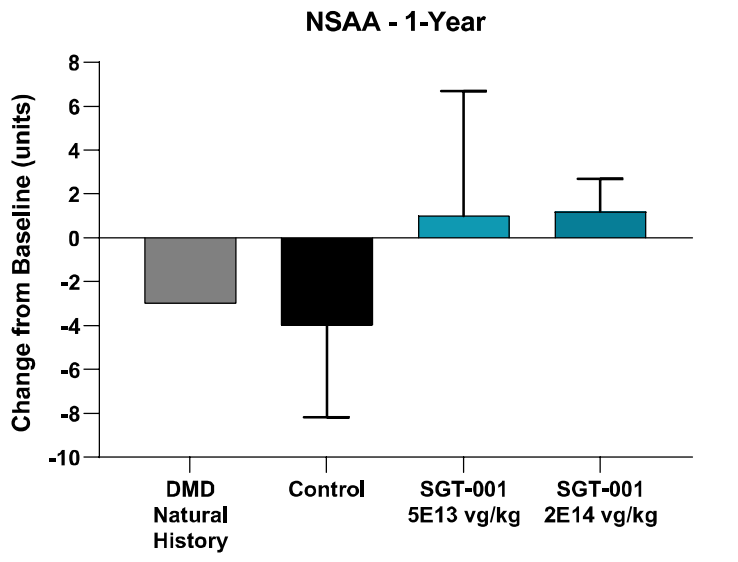
NSAA - 1-Year
Change from Baseline (units)
8
6
4
2
0
-2
-4
-6
-8
-10
DMD Natural History
Control
SGT-001 5E13 vg/kg
SGT-001 2E14 vg/kg

-3.0 unit expected decline in 12 months after age 6.3 (Muntoni et al 2019)

Conclusions

- Patients administered SGT-001 on average showed improved spontaneous walking velocity at 1-year post-treatment as measured by the SV95C assessment through the ActiMyo wearable device
 - Results indicate improved maximal performance in SGT-001 treated patients
- These improvements in SV95C align with change from baseline results in the 6MWT and NSAA clinical evaluations of ambulatory function
 - Stable or improved 6MWT distances and NSAA scores in SGT-001 treated patients
- Across all 3 evaluations of ambulatory function, SGT-001 treated patients show a clinical course that diverges from the characterized natural history declines that would be expected over a 1-year period
- SV95C assessment through the ActiMyo wearable device serves as a meaningful, objective endpoint for the evaluation of ambulatory function that may provide additional sensitivity in evaluating the therapeutic efficacy of clinical drug candidates for DMD